JAK/STAT signaling pathway negatively regulates angiotensin II AT_1 receptor induced cardiac hypertrophy in rat cardiomyocytes

Bing Hui Wang[1], Ross Hannan[2], Lawrence S. Zisman[3], Shreefal Mehta[3], Stephen Su[1], Christopher Burns[1], Xianyoung Bu[1], Max Joffe[1], Yue Wang[1], Irma Kruszelnicki[1], Emmanuelle Fantino[1], Claire Mitchell[1], Ian Phillips[1] ,Andrew F. Wilks[1], Walter Thomas[4]

1. Cytopia Pty Ltd, Commercial Road, Melbourne, VIC 3004 Australia 2. The Peter MacCallum Cancer Institute, St Andrews Place, East Melbourne, VIC 3002 Australia

3. Myomatrix Therapeutics, LLC. One University Place, DB-224, Rensselaer NY 12144 USA 4. Baker Heart Research Institute, Commercial Road, Melbourne, VIC 3004 Australia

INTRODUCTION

The pleiotropic effects of angiotensin II (Ang II) play important roles in various cardiac functions, including cardiac growth (hypertrophy), through the G-protein-coupled AT_1 receptor and stimulate cardiac growth by activation of protein kinases such as protein kinase C (PKC), and MAP kinases. Cardiac hypertrophy in response to increased cardiac demand is of benefit initially, but compensatory hypertrophy may transition to heart failure over time. ACE inhibitors and antagonists of AT_1 receptors have demonstrated that blocking chronic activation of AT_1 receptors in the heart ameliorates heart failure.

Several lines of evidences show that Ang II activates the JAK/STAT pathway directly through its G-protein-coupled AT_1 receptor. However, the precise physiological role of the JAK/STAT signaling pathway has not been elucidated in cardiomyocytes. In particular, some studies are confounded by the use of partially-selective JAK2 inhibitors (eg, AG490) that also inhibit the EGF receptor (EGFR) and other kinases.

Prior studies have shown that Ang II stimulates cardiac growth through transactivation of EGFR. In this study, we examined the role of the JAK/STAT pathway on Ang II stimulated cardiac growth and its potential role in cardiac hypertrophy in rat neonatal cardiomyocytes (RCMs).

METHODS

CYC Compounds

All CYC compounds used in this study were synthesized by Cytopia's Chemists and purified to greater than 95% by HPLC.

Protein Kinase Assays

Protein kinases were cloned, and the kinase domain-GST fusion protein expressed in Baculovirus and purified for kinase screens. The enzyme assays were performed with ALPHA (Amplified Luminescence Proximity Homogeneous Assay) screen (PerkinElmer) using peptide substrates selective for respective kinases.

Cell Proliferation Assay

The activity and selectivity of CYC compounds was determined by *in vitro* cell proliferation assays using cytokine dependent cell lines obtained from ATCC (CTLL-2, an Il-2-dependent mouse T lymphocyte; TF-1, a granulocytic-marcrophage colony-stimulating factor (GMCSF)-dependent human erythroleukaemia cell line).

An IL-3-dependent mouse myeloid cell line, Baf3 (from ATCC), was engineered to express the kinase domain of JAK2 or JAK3 (TEL-JAK2 or TEL-JAK3). The lines became IL-3-independent, but dependent upon the respective kinase activity to grow. IL-2 is reported to signal through JAK1 and JAK3, whereas GM-CSF is reported to signal through JAK2. CYC compounds were screened for their capacity to inhibit cell proliferation (Alamar Blue uptake) and thereby JAK2/3. Cell density was $1x10^4$ cells/ml (96-well plates) in the presence of 10 ng/mL of IL-2 or GM-CSF for CTLL-2 and TF-1, respectively. Baf3/TEL-JAK2 and Baf3/TEL-JAK3 were grown in the absence of growth factor. 72 hours after adding CYC compounds, Alamar Blue was added and cells incubated for an additional 8 to 16 hours. Fluorescent intensity was measured by a FLUOstar Reader at emission of 590 nm and excitation of 544 nm light.

Cardiac Myocytes Isolation and Culture

Cardiomyocytes were isolated from ventricles of 1-day-old Sprague-Dawley rat pups and plated in 12 well-plates. After 24 hours incubation, myocytes were infected with purified AT_1-receptor (AT_1R) adenovirus at a multiplicity of infection (MOI) of 10-20. At this level of infectivity, receptor expression was aboout 300-500 fmol/mg protein and was associated with robust Ang II-mediated hypertrophy.

Induction of Hypertrophy

One day after infection with adenovirus, AT_1R mediated hypertrophy was initiated by adding Ang II (100 nmol/L). After 72 hours of stimulation, cells were harvested and hypertrophy defined as a significant increase in protein content (Lowry assay) in the absence of any significant change in DNA content (Burton assay). Cardiac hypertropy (growth) was determined by protein to DNA ratios.

Western Blot Analysis

For Western Blot analysis, myocytes were treated with compounds for 20 min and stimulated with Ang II for 10 min before cells were lysed with RIPA buffer. 15 µg protein from the cleared lysates were loaded on SDS-PAGE and Western blotted with antibodies, including anti-pY^{705}-STAT3, pY^{694}-STAT5 and pY^{204}/T^{202} p42/p44 (Cell Signaling Technology) were performed.

Quantitative RT-PCR

For the ANF gene expression assay, cultured rat neonatal cardiac myocytes were treated for 48 hours in serum free media plus 10 µM Ang II, or vehicle, with and without a selective JAK2 inhibitor. Total mRNA was extracted with RNA-STAT 60. RT-qPCR was performed with Syber Green to measure ANF and GAPDH. Relative gene expression was measured with the cycle threshold method. Each condition was performed in triplicate. Statistical analysis was performed with ANOVA and the Bonferroni correction.

Table 1 Inhibition of protein Kinase activity by selective JAK inhibitors (IC_{50} nM)

Compound	JAK1	JAK2	JAK3	Tyk2	Other kinases	Selectivity (fold)
10325	15	1.25	40	1	>1000	≥800
10478	ND	400	5000	ND	>7500	>18
10647	ND	60	100	ND	>1250	>20
10679	ND	40	2500	ND	>2500	>60
10977	ND	>10000	30	ND	>10000	>330
11069	112	15	10	ND	>10000	≥100

* Selectivity fold vs kinases other than JAKs.

The inhibitory activity of CYC compounds on 20 protein kinases including JAK kinases was tested with ALPHAscreen.

As shown in Table 1, compound 10325 inhibited all four JAKs and therefore is a *pan*-JAK inhibitor. In contrast, 10647 and 11069 are selective inhibitors of JAK2 and JAK3; 10478 and 10679 are selective JAK2 inhibitor; 10977 is a highly selective JAK3 inhibitor.

These compounds had similar selectivity (Table 2) in cell-based proliferation assays.

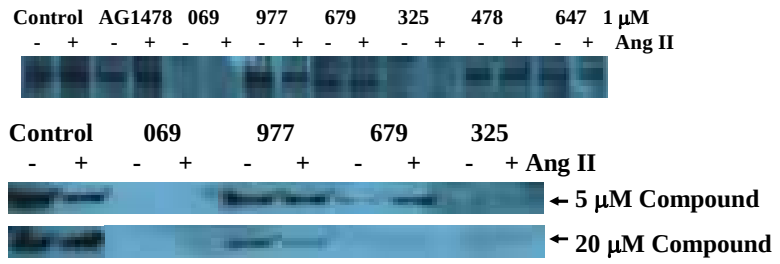

Control AG1478 069 977 679 325 478 647 1 µM
- + - + - + - + - + - + - + - + Ang II
Control 069 977 679 325
- + - + - + - + - + Ang II
5 µM Compound
20 µM Compound

Figure 1 Cell lysates of RCMs overexpressing the AT1R and treated with varying doses of Ang II were subjected to Western blot with the anti-Y^p STAT3 antibody.

STAT3 was constitutively phosphorylated at tyrosine (705) and its phosphorylation inhibited by the *pan*-JAK and JAK2/3 inhibitors, compound 325 and 069 respectively. Moreover, a highly selective JAK3 inhibitor, compound 977, had little effect on STAT3 tyrosine phosphorylation whereas the selective JAK2 inhibitor 679 blocked STAT3 phosphorylation at higher concentration. These data indicate that STAT3 activation in RCM is likely *via* JAK2. The selective EGFR inhibitor, AG1478, did not effect STAT3 phoshporylation in RCM overexpressing the AT1R.

(key: -, no Ang II; +, in presence of Ang II.)

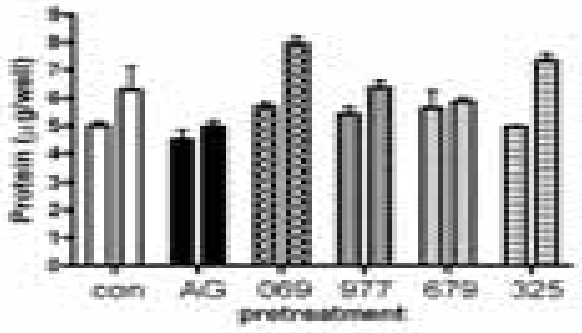

Protein (µg/well)
con AG 069 977 679 325
pretreatment

Figure 2 Ang II induced RCM hypertrophic growth through transactivation of the EGFR and was inhibited by the EGFR inhibitor, AG1478. In contrast, RCMs pre-treated with the *pan*-JAK inhibitor (325) and JAK2/3 inhibitor (069) enhanced RCM hypertropic growth in response to Ang II stimulation as measured by protein/DNA ratios while neither the selective JAK3 inhibitor 977 nor the selective JAK2 inhibitor 679 altered RCM growth stimulated by Ang II relative to controls. Thus, these results demonstrated that simultaneous inhibition of both JAK2 and JAK3 potentiated Ang II stimulated cardiac hypertrophy whereas inhibition of either JAK2 or JAK3 did not (assuming you have p values and I am interpreting the figure correctly).

Table 2 Inhibition of cell proliferation by selective JAK inhibitors (IC_{50} µM)

Compound	TEL-JAK2	TEL-JAK3	CTLL-2	TF-1	Others cell lines tested
10325	0.5	2	0.1	0.5	>20
10478	25	>200	>150	20	>200
10647	4	8	3	5	>8
10679	0.6	3	>20	4	>10
10977	5	0.4	2	>20	>20
11069	2	0.8	0.25	1.25	>10

Wally, Would you mind write some thing for the results here that I Have some trouble to describe it clearly. Thanks. Bing

Myocytes pictures from Wally to be added in.

As shown in this Figure, Ang II induced RCM hypertrophic growth through transactivation of EGFR and was inhibited by EGFR inhibitor, AG1478, as have been demonstrated previously and shown above.

RCMs (overexpressing the AT1R?) pre-treated with *pan*-JAK inhibitor 325 and JAK2/ 3 inhibitor 069, enhanced RCM hypertropic growth stimulated by Ang II (what uM concentration?).

The selective JAK3 inhibitor 977 did not affect RCM growth stimulated by Ang II. What was the effect of 647 and 679 on Ang II induced RCM growth?

Thus, these results demonstrated that JAK2/STAT3 activation might play a negative regulation role on RCM growth stimulated by Ang II. Interestingly, RCM do not grow well in the present of high concentrations of JAK inhibitors particularly selective JAK2 inhibitor 679 but such effect can be "rescued" by Ang II. Thus, JAK/STAT pathway negatively regulates Ang II/AT_1R induced RCM growth. Furthermore, JAK2/STAT3 pathway may have some maintenance effect in RCM growth as well.

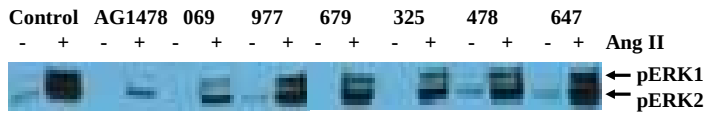

Control AG1478 069 977 679 325 478 647
- + - + - + - + - + - + - + - + Ang II
pERK1
pERK2

Figure 4. EGFR stimulated cell growth by activating MAP kinase ERK1/2. The selective EGFR inhibitor, AG1478, inhibits MAP kinase, ERK1 and 2, activation induced by Ang II. But all the JAK kinase inhibitors tested have no effects on MAPK activation induced by Ang II. This suggested that activation of MAPK induced RCM hypertrophic growth by Ang II, and that this occured *via* transactivation of the EGF receptor by the AT_1 receptor. Neither non-selective nor selective JAK inhibitors effected ERK1 or 2 phosphorylation.

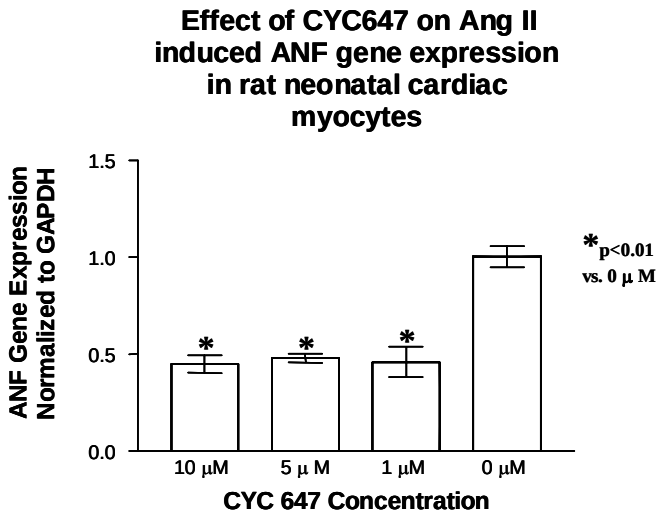

Effect of CYC647 on Ang II induced ANF gene expression in rat neonatal cardiac myocytes
ANF Gene Expression Normalized to GAPDH
1.5
1.0
0.5
0.0
10 µM
5 µ M
1 µM
0 µM
CYC 647 Concentration
*p<0.01 vs. 0 µ M

In the presence of 1, 5, or 10 µM CYC647, ANF gene expression normalized to GAPDH was reduced by over 50% compared to that in cells treated with Ang II alone. In contrast, the *pan*-JAK inhibitor 325 potentiated Ang II induced ANF gene expression (Ang II 10 µM +CYC325 1 µM ANF gene expression 4.08 relative to Ang II 10 µM alone).

RESULTS

- A panel of selective JAK inhibitors, CYC compounds, were tested for their capacity to inhibit Ang II AT_1 receptor activation of JAK/STATs and effects on EGFR transactivation.
- Selective JAK inhibitors inhibited STAT3 activation, both basally and activated by Ang II, but they did not affect the activation of MAPK *via* transactivation of EGFR.
- Non-selective JAK inhibitors enhanced myocyte hypertrophy as measured by protein/DNA ratios and ANF gene expression induced by Ang II.
- A Selective JAK2 inhibitor decreased ANF expression induced by Ang II.
- However, selective JAK inhibitors did not inhibit Ang II stimulated MAPK activation, indicating that the JAK/STAT pathway is independent of the EGF receptor.

CONCLUSIONS

- This study demonstrated that selective inhibition of JAK2 decreased Ang II stimulated ANF gene expression (a marker of heart failure).
- Selective inhibition of JAK2/STAT signaling may block the progression to a heart failure phenotype.